1789

Presented at the

65th ASH Annual Meeting & Exposition

San Diego, CA, USA • December 9-12, 2023

ALK2 and JAK2 Inhibition for Improved Treatment of Anemia in Myelofibrosis Patients: Preclinical Profile of an ALK2 Inhibitor Zilurgisertib in Combination With Ruxolitinib

Matthew C. Stubbs, Michelle Pusey, Xiaoming Wen, Katherine Drake, Nina Zolotarjova, Amanda Smith, Maryanne Covington, Guofeng Zhang, Ricardo Macarrón, Sunkyu Kim

Incyte Research Institute, Wilmington, DE, USA

Abstract

Introduction: Anemia is common in patients with myelofibrosis (MF) and is associated with the need for red blood cell transfusion, and poor clinical prognosis. Janus kinase (JAK) inhibitors such as ruxolitinib are extensively utilized to treat the symptoms of MF and improve quality of life, but are also broadly myelosuppressive. New therapeutic interventions that can block JAK signaling while avoiding anemia would benefit patients with MF by reducing unwanted side effects that lead to discontinuation and/or suboptimal dosing. Recent studies indicate that inhibition of activin A receptor 1 (ACVR1)/activin receptor-like kinase 2 (ALK2), a bone morphogenetic protein (BMP) receptor upstream of hepcidin transcriptional regulation, could reduce serum hepcidin levels in patients with MF and improve anemia.[1,2] Reducing levels of hepcidin, a key hormone in regulating iron homeostasis, and restoring erythropoiesis would benefit patients with MF being treated with JAK inhibitors. Currently, several options for ALK2 inhibition are approved or under study, including momelotinib (JAK1/JAK2 and ALK2 inhibitor) and pacritinib (JAK2, interleukin-1 receptor-associated kinase 1 [IRAK1], and ALK2 inhibitor). Recent clinical data from momelotinib and pacritinib studies indicate ALK2 inhibition improves anemia in patients with MF. We therefore sought an ALK2 inhibitor that is potent, selective, and could be dose titrated. Here we discuss zilurgisertib, an ALK2-specific inhibitor that could be dose titrated to fit patient needs.

Methods and Results: In biochemical and cellular assays, zilurgisertib inhibited ALK2 kinase activity and SMAD1/5 phosphorylation, with half-maximal inhibitory concentration (IC_{50}) values of 11 nM and 69 nM, respectively. In Huh7 cells stimulated with BMP-6, zilurgisertib inhibited hepcidin production with an IC_{50} of 20 nM, demonstrating the compound is a potent ALK2 inhibitor capable of regulating hepcidin. To assess possible off-target effects of zilurgisertib, kinome profiling at 100 µM adenosine triphosphate (ATP) was performed on each ALK2 inhibitor at Reaction Biology (Malvern, PA, USA), to determine the overall specificity of these inhibitors across 356 kinases. At 200 nM, zilurgisertib only inhibited ALK2, ALK1 (to 50%), and ALK6 (to 48%). Further, 20 µM zilurgisertib did not affect viability of human embryonic kidney 293 (HEK293) cells, a human cell line commonly used to assess general cell health and compound toxicity. Likewise, zilurgisertib did not affect viability of human fibroblasts or endothelial cells at concentrations up to 5 µM.

We next explored whether the combination of ruxolitinib and zilurgisertib could replicate effects reported for momelotinib[1] and pacritinib[2] by suppressing hepcidin and restoring erythropoiesis in an in vivo mouse model of cancer-induced anemia. B16F10 cells were injected intraperitoneally, mimicking a metastatic tumor that leads to anemia 1 week after injection. Zilurgisertib dose dependently improved hemoglobin (by 2-3 g/dL) and red blood cell counts, while reducing both liver phosphorylated SMAD (pSMAD) and circulating hepcidin levels by ≥50% vs vehicle control. Combination with ruxolitinib did not alter this activity, indicating that inhibition of JAK2 activity should not impede erythropoiesis prompted by ALK2 inhibition.

Conclusion: Taken together, the potency and selective on-target activity of zilurgisertib suggest that ALK2 inhibition could reduce hepcidin and improve anemia, and that combination of zilurgisertib with ruxolitinib is a rational and attractive approach. The combination of ruxolitinib and zilurgisertib is currently in a phase 1 clinical trial in patients with anemia due to myeloproliferative disorders (NCT04455841).

Zilurgisertib Selectively Inhibits ALK2 in Biochemical Assays

ALK2 Inhibitor	Biochemical Potency (IC_{50} nM) at 100 µM ATP			
	ALK1	ALK2	ALK3	ALK5
Zilurgisertib	155	11	1135	>10,000
Momelotinib	1545	270	>10,000	>10,000
Pacritinib	131	88	>10,000	>10,000

ALK, activin receptor-like kinase; ATP, adenosine triphosphate; IC_{50}, half-maximal inhibitory concentration.

- Biochemical kinase activity was evaluated at 100 µM ATP using the LANCE® Ultra Kinase Assay (PerkinElmer, Waltham, MA, USA) to measure phosphorylation of ULight™-DNA Topoisomerase 2-alpha peptide
- All data represent geometric mean values from repeated assays (n=3-8)

Zilurgisertib Potently Inhibits ALK2 Signaling Pathway in Cell-Based Assays

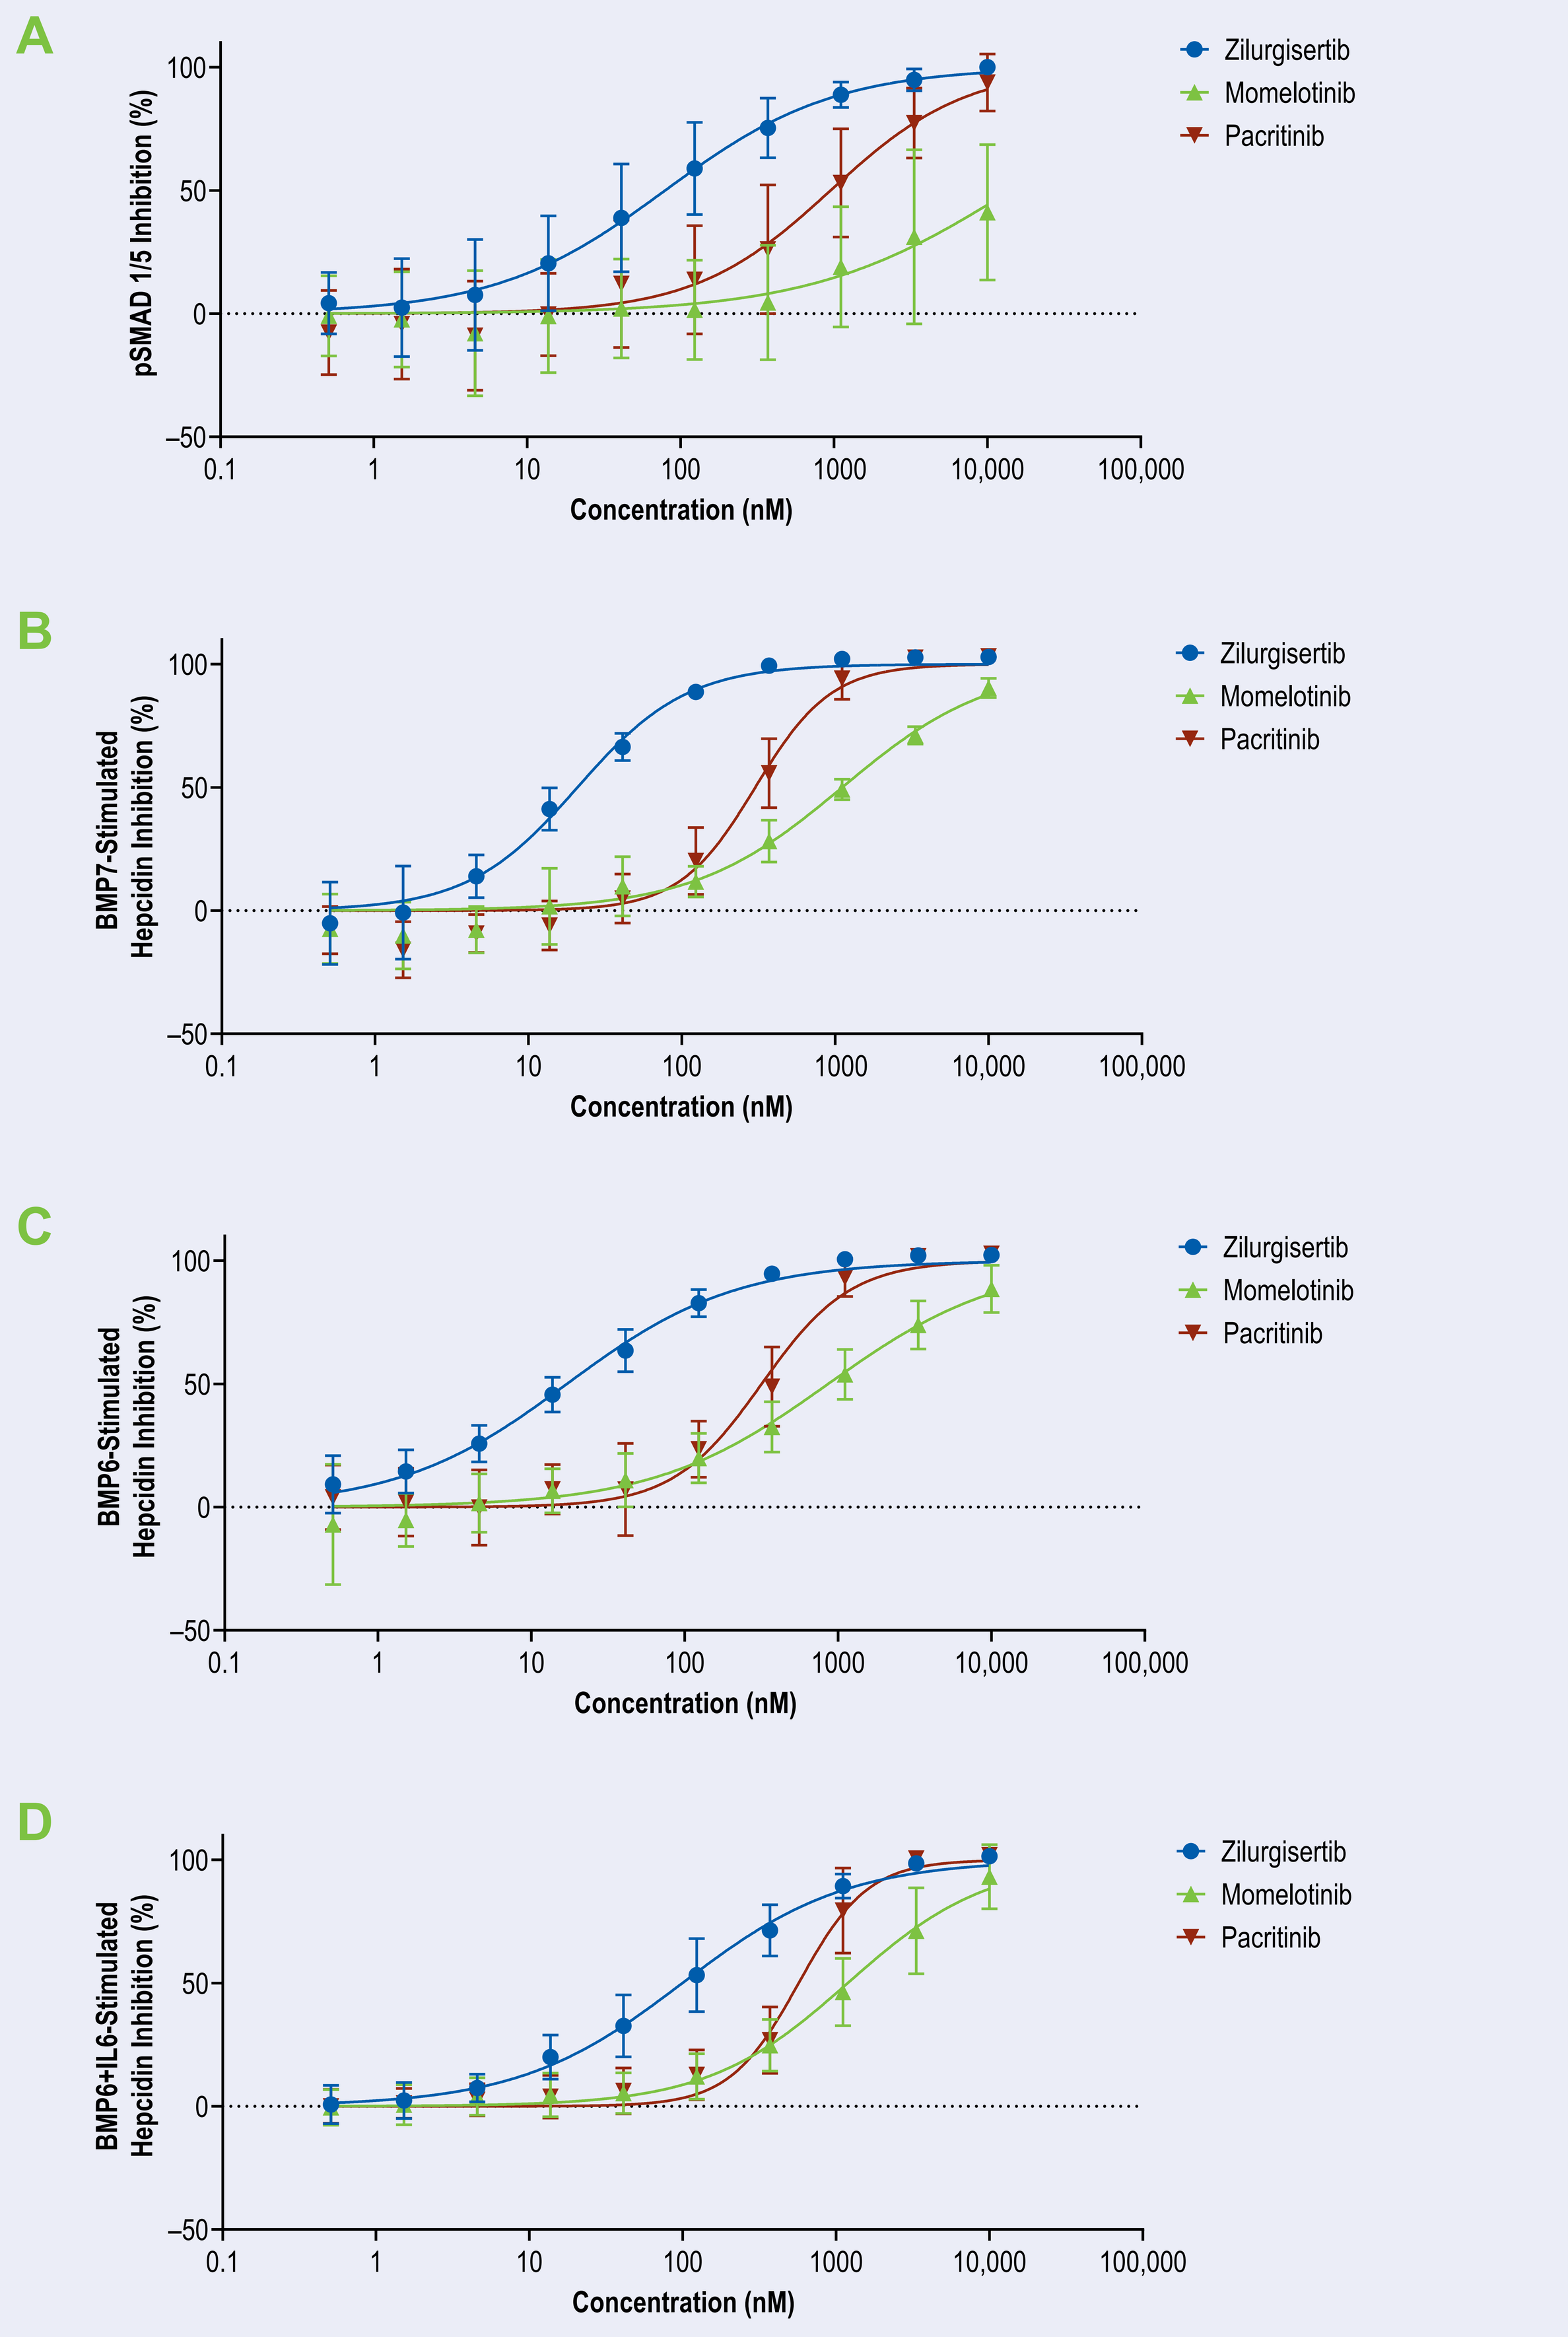
A
B
C
D
pSMAD 1/5 Inhibition (%)
BMP7-Stimulated Hepcidin Inhibition (%)
BMP6-Stimulated Hepcidin Inhibition (%)
BMP6+IL6-Stimulated Hepcidin Inhibition (%)
Concentration (nM)
Zilurgisertib
Momelotinib
Pacritinib

Data represent mean values from repeated assays ± standard deviations. ALK, activin receptor-like kinase; BMP, bone morphogenetic protein; IL, interleukin; pSMAD, phosphorylated SMAD.

ALK2 Inhibitor	Cell-Based Assay			
	pSMAD 1/5 IC_{50} (nM)	Hepcidin IC_{50} (nM)	Huh7 BMP6+IL6 Hepcidin IC_{50} (nM)	Huh7 BMP6 Hepcidin IC_{50} (nM)
Zilurgisertib	69	20	99	17
Momelotinib	4709	1081	1213	863
Pacritinib	698	294	543	301

ALK, activin receptor-like kinase; BMP, bone morphogenetic protein; IL, interleukin; pSMAD, phosphorylated SMAD.

A. pSMAD1 levels were measured in HeLa cells after 1 hour pretreatment with each ALK2 inhibitor and 30 minutes stimulation with BMP7
B. Hepcidin levels were measured in Huh7 cells after 24 hours of treatment with each ALK2 inhibitor and BMP7
C. Hepcidin levels were measured in Huh7 cells after 24 hours of treatment with each ALK2 inhibitor and BMP6
D. Hepcidin was induced through both BMP-SMAD and IL6-JAK-signal transducer and activator of transcription (STAT) pathways in Huh7 cells and hepcidin levels were measured after 24 hours of treatment with each ALK2 inhibitor

Selectivity of Zilurgisertib for ALK2 Over Other Kinases

Zilurgisertib

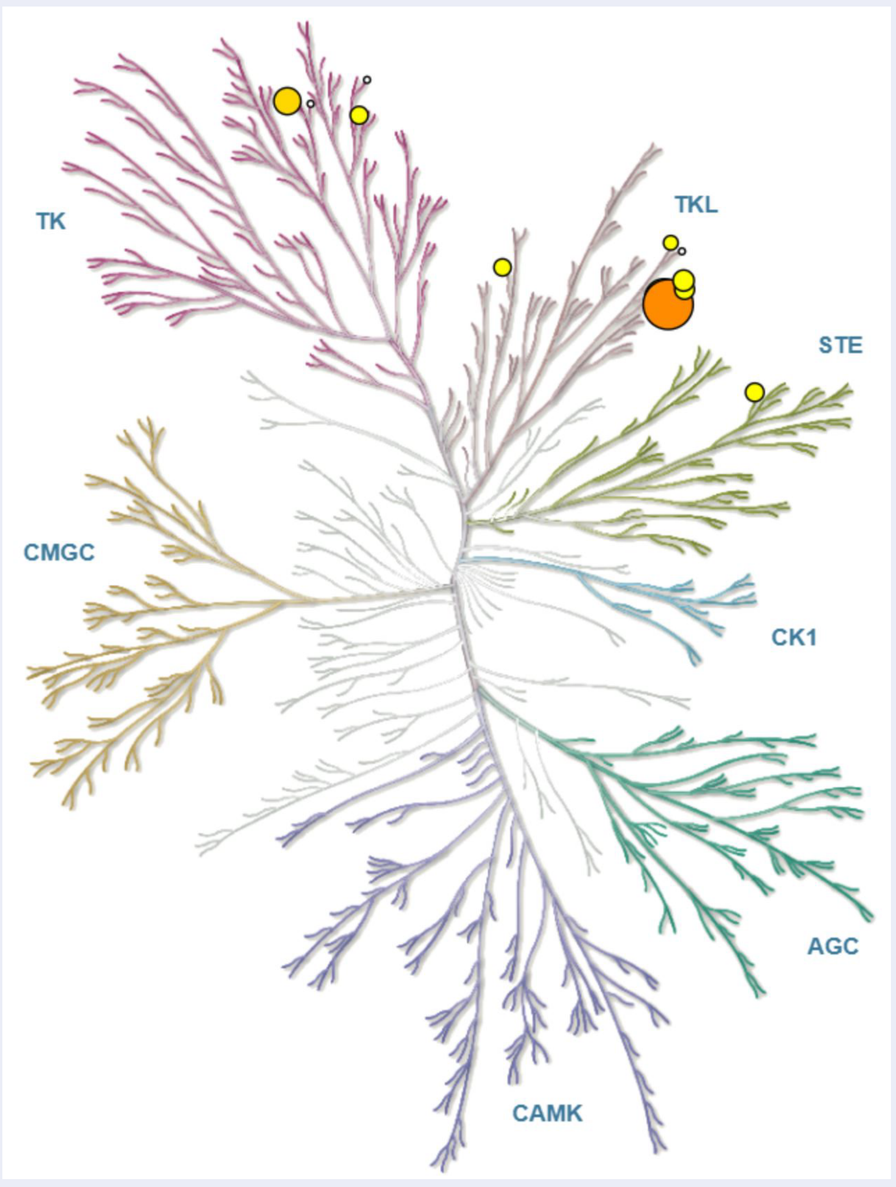
TK
TKL
STE
CMGC
CK1
AGC
CAMK

Momelotinib

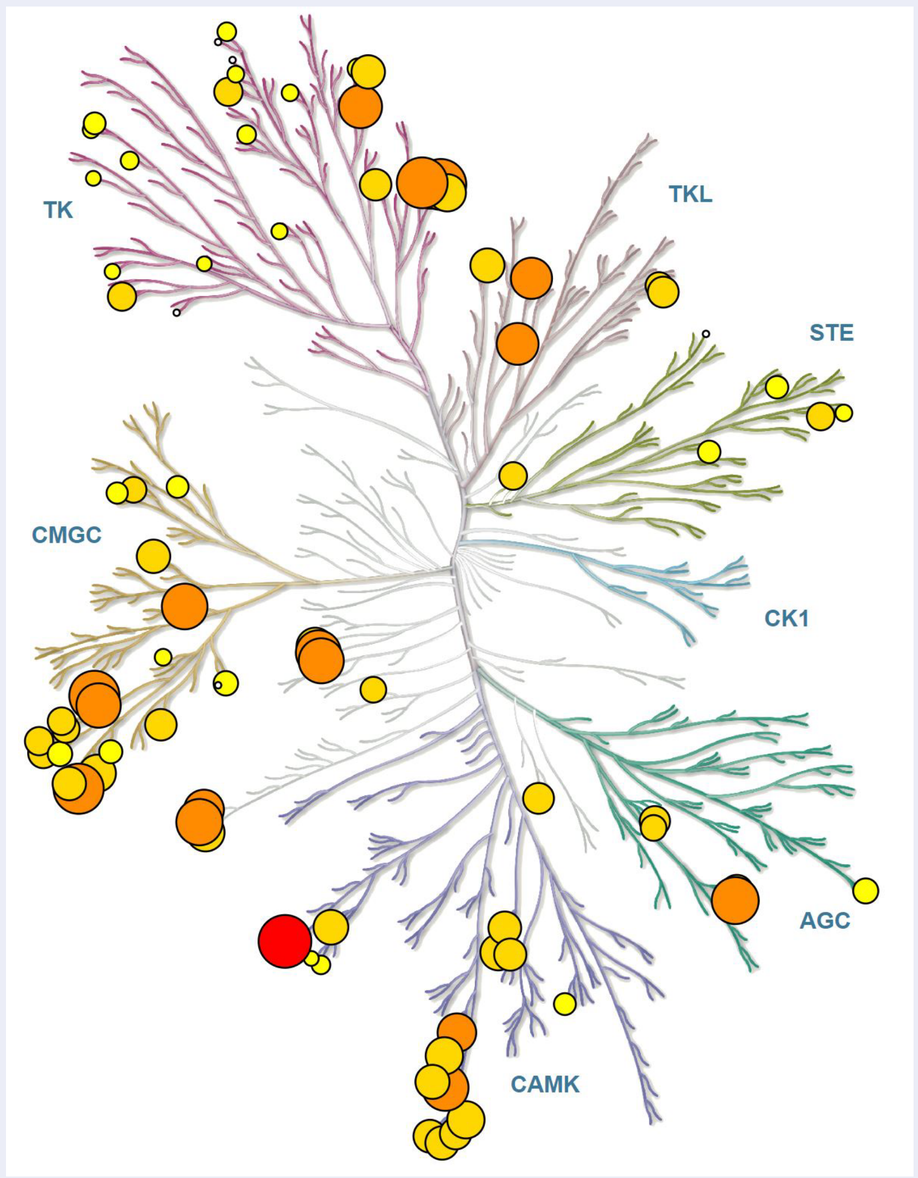
TK
TKL
STE
CMGC
CK1
AGC
CAMK

Pacritinib

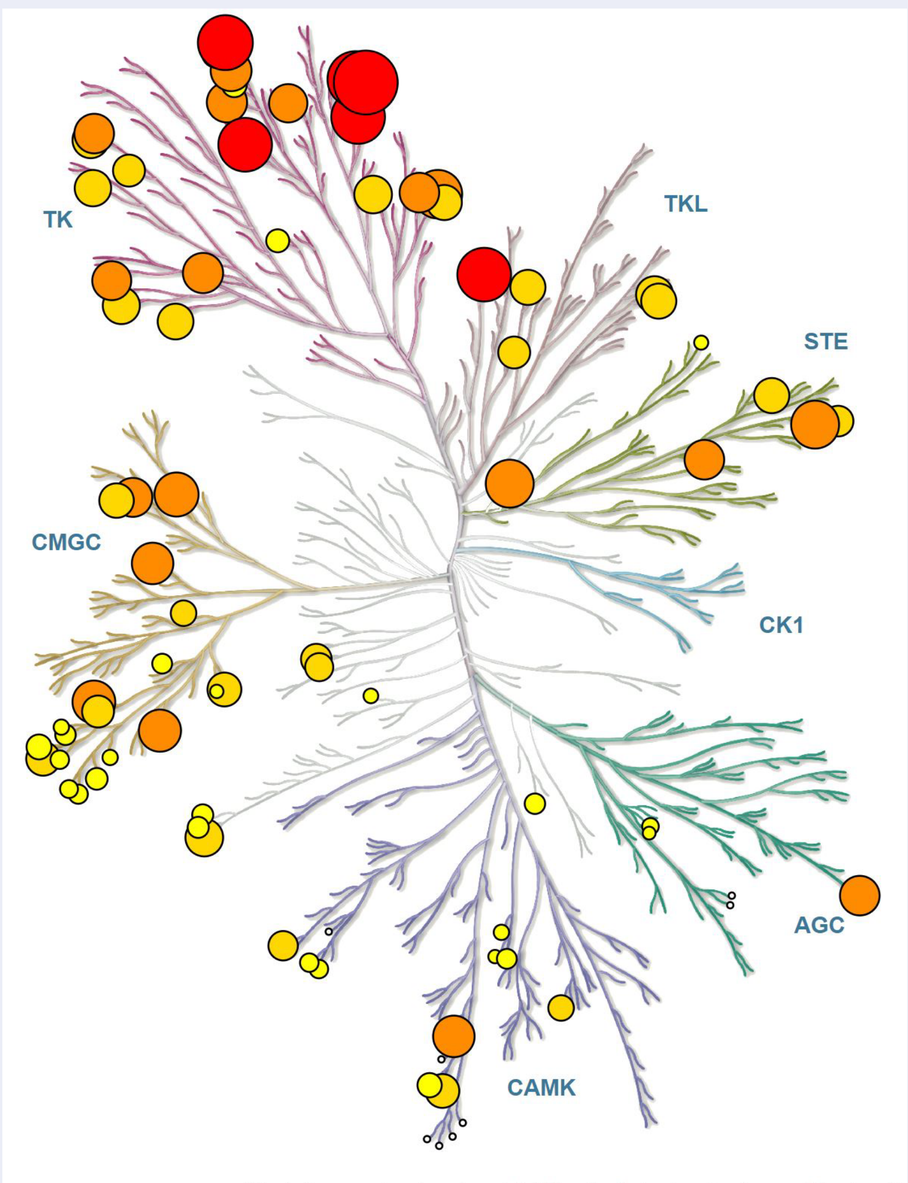
TK
TKL
STE
CMGC
CK1
AGC
CAMK

IC_{50}
- <10 nM
- <100 nM
- <1000 nM
- <10,000 nM
- ≥10,000 nM

Size by IC_{50} value

Illustrations reproduced courtesy of Cell Signaling Technology, Inc. (www.cellsignal.com). IC_{50}, half-maximal inhibitory concentration.

- Possible off-target effects of zilurgisertib and other ALK2 inhibitors across 356 kinases were assessed by kinome profiling
- Assays run at 100 µM ATP; performed by Reaction Biology (Malvern, PA, USA)
- At 200 nM, zilurgisertib only inhibited ALK2, ALK1 (to 50%), and ALK6 (to 48%)

Zilurgisertib Exhibits Minimal Off-Target Mediated Growth Inhibition

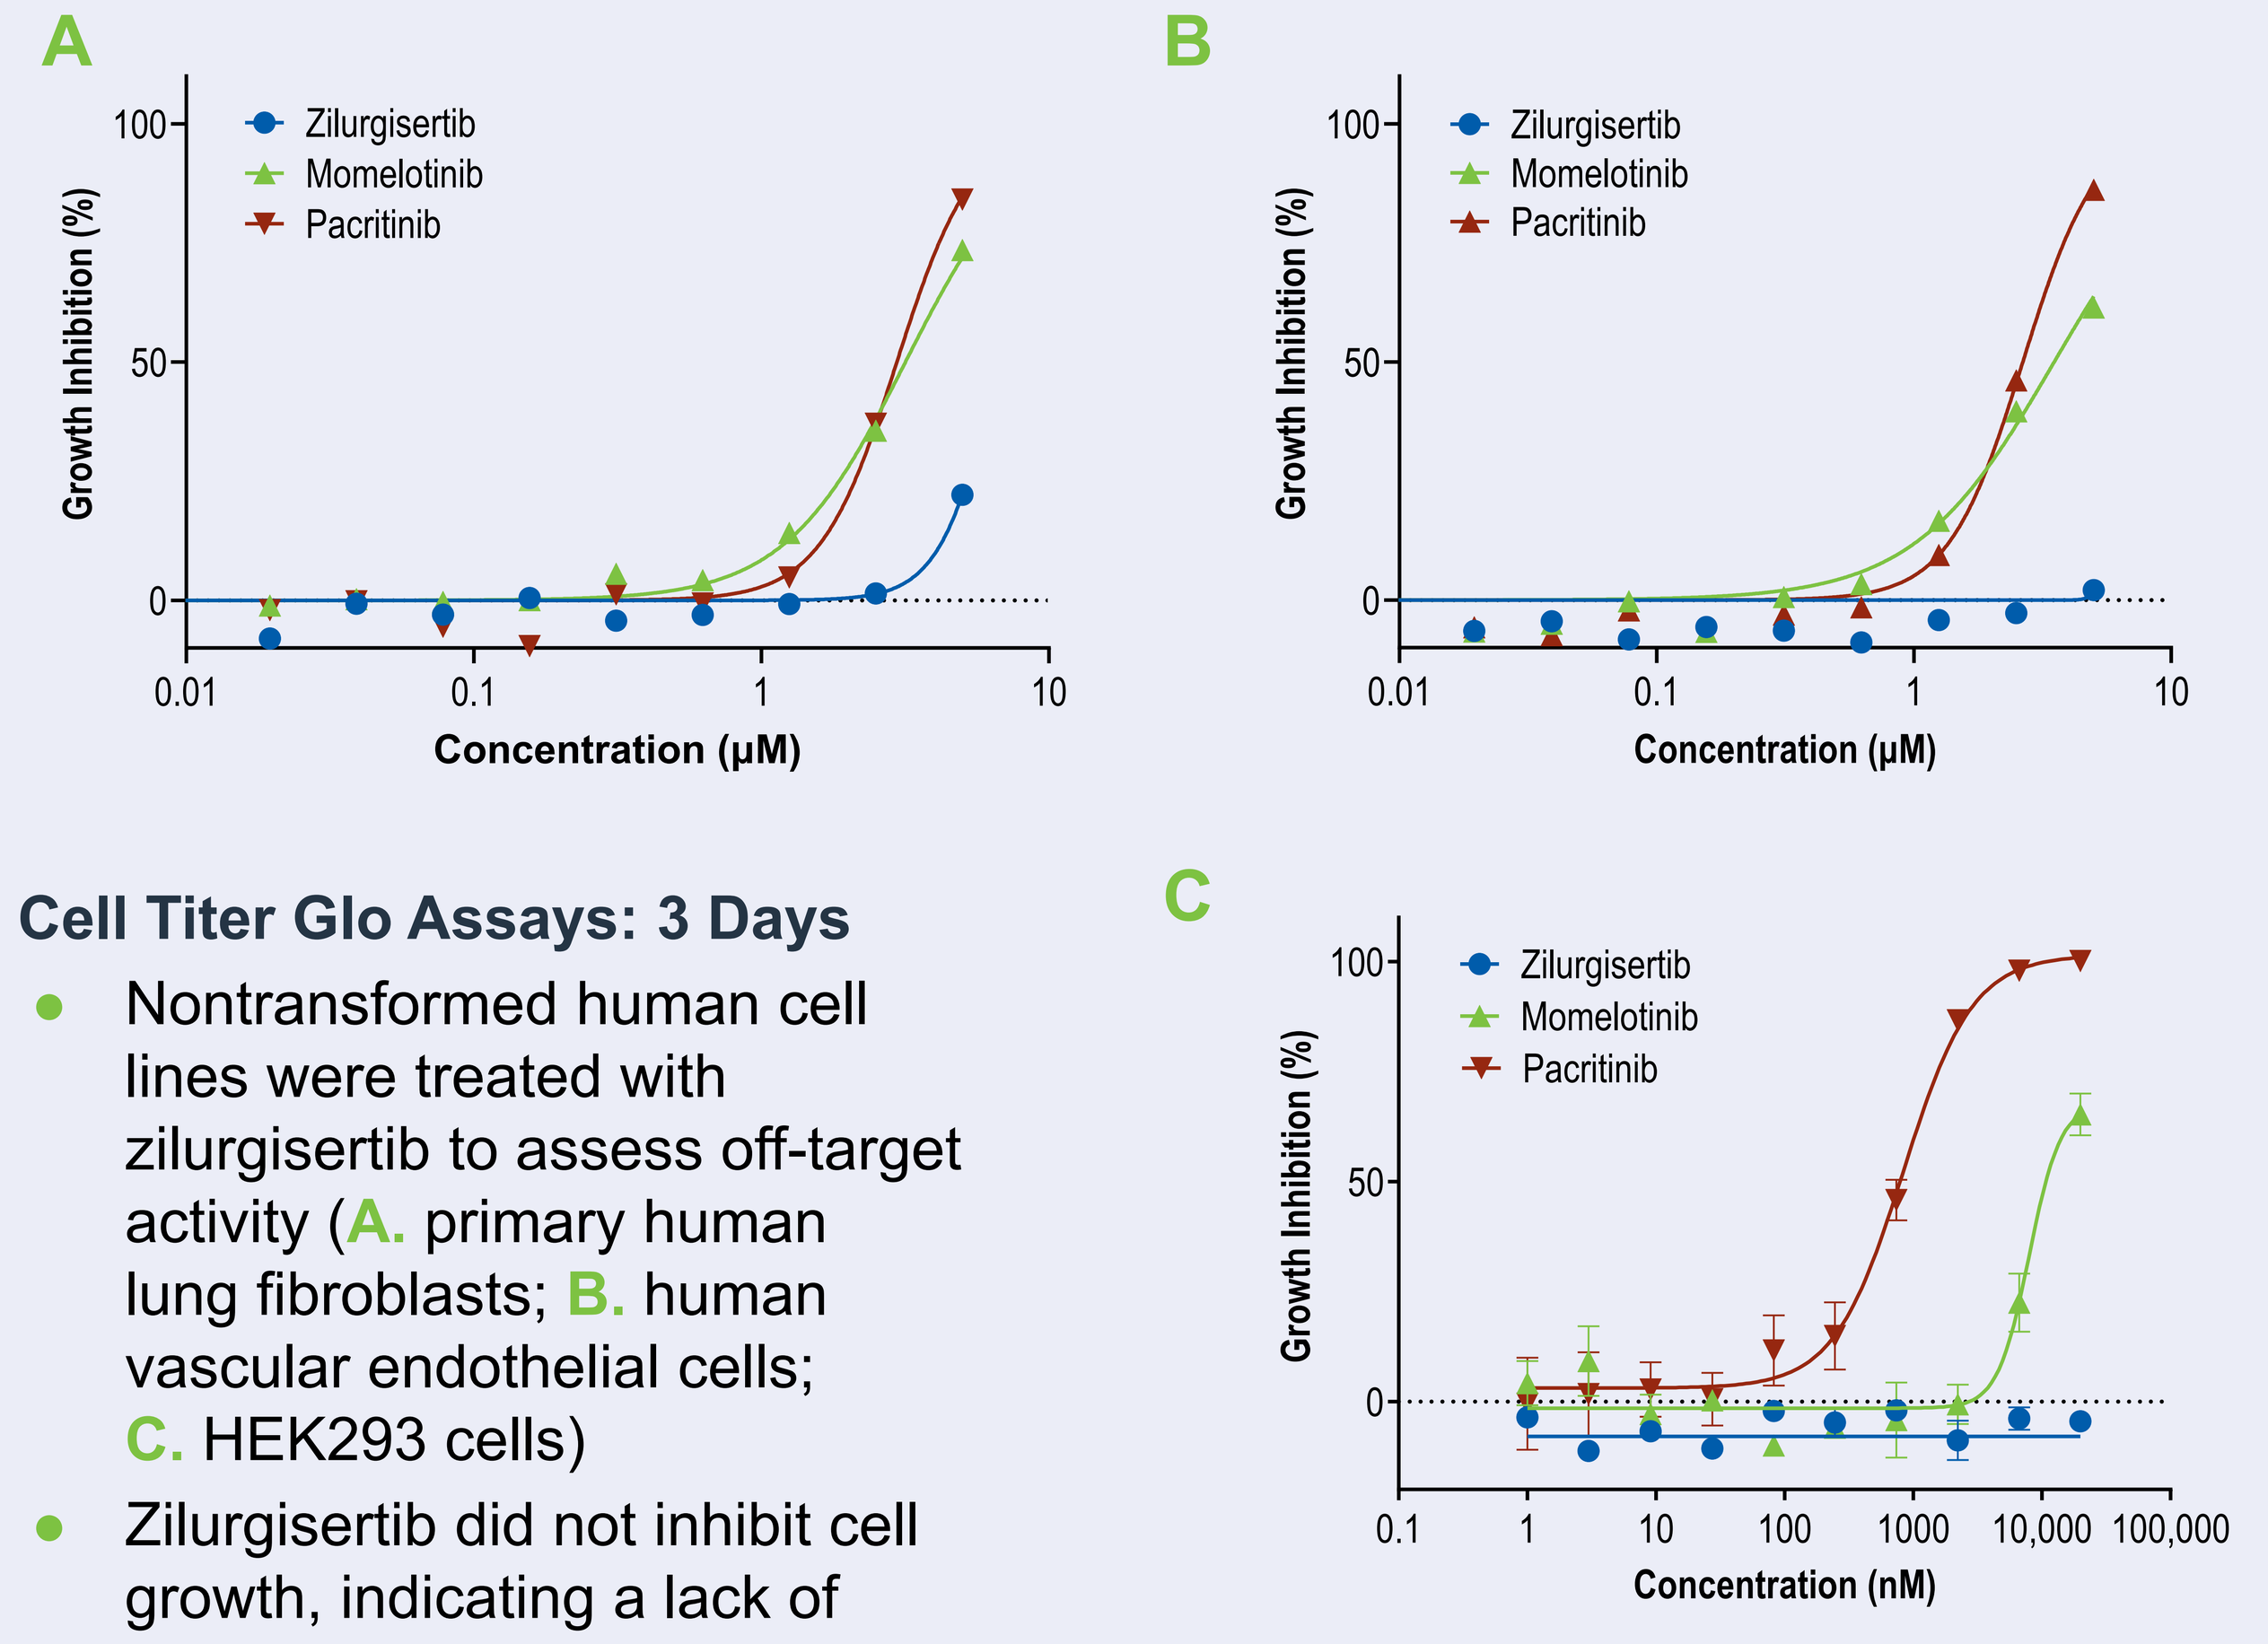
A
B
C
Growth Inhibition (%)
Concentration (µM)
Concentration (nM)
Zilurgisertib
Momelotinib
Pacritinib

Cell Titer Glo Assays: 3 Days

- Nontransformed human cell lines were treated with zilurgisertib to assess off-target activity (**A.** primary human lung fibroblasts; **B.** human vascular endothelial cells; **C.** HEK293 cells)
- Zilurgisertib did not inhibit cell growth, indicating a lack of off-target activity

Data represent mean from repeated assays (± standard deviation in C).

Zilurgisertib Shows Efficacy in a Murine Cancer-Induced Anemia Model

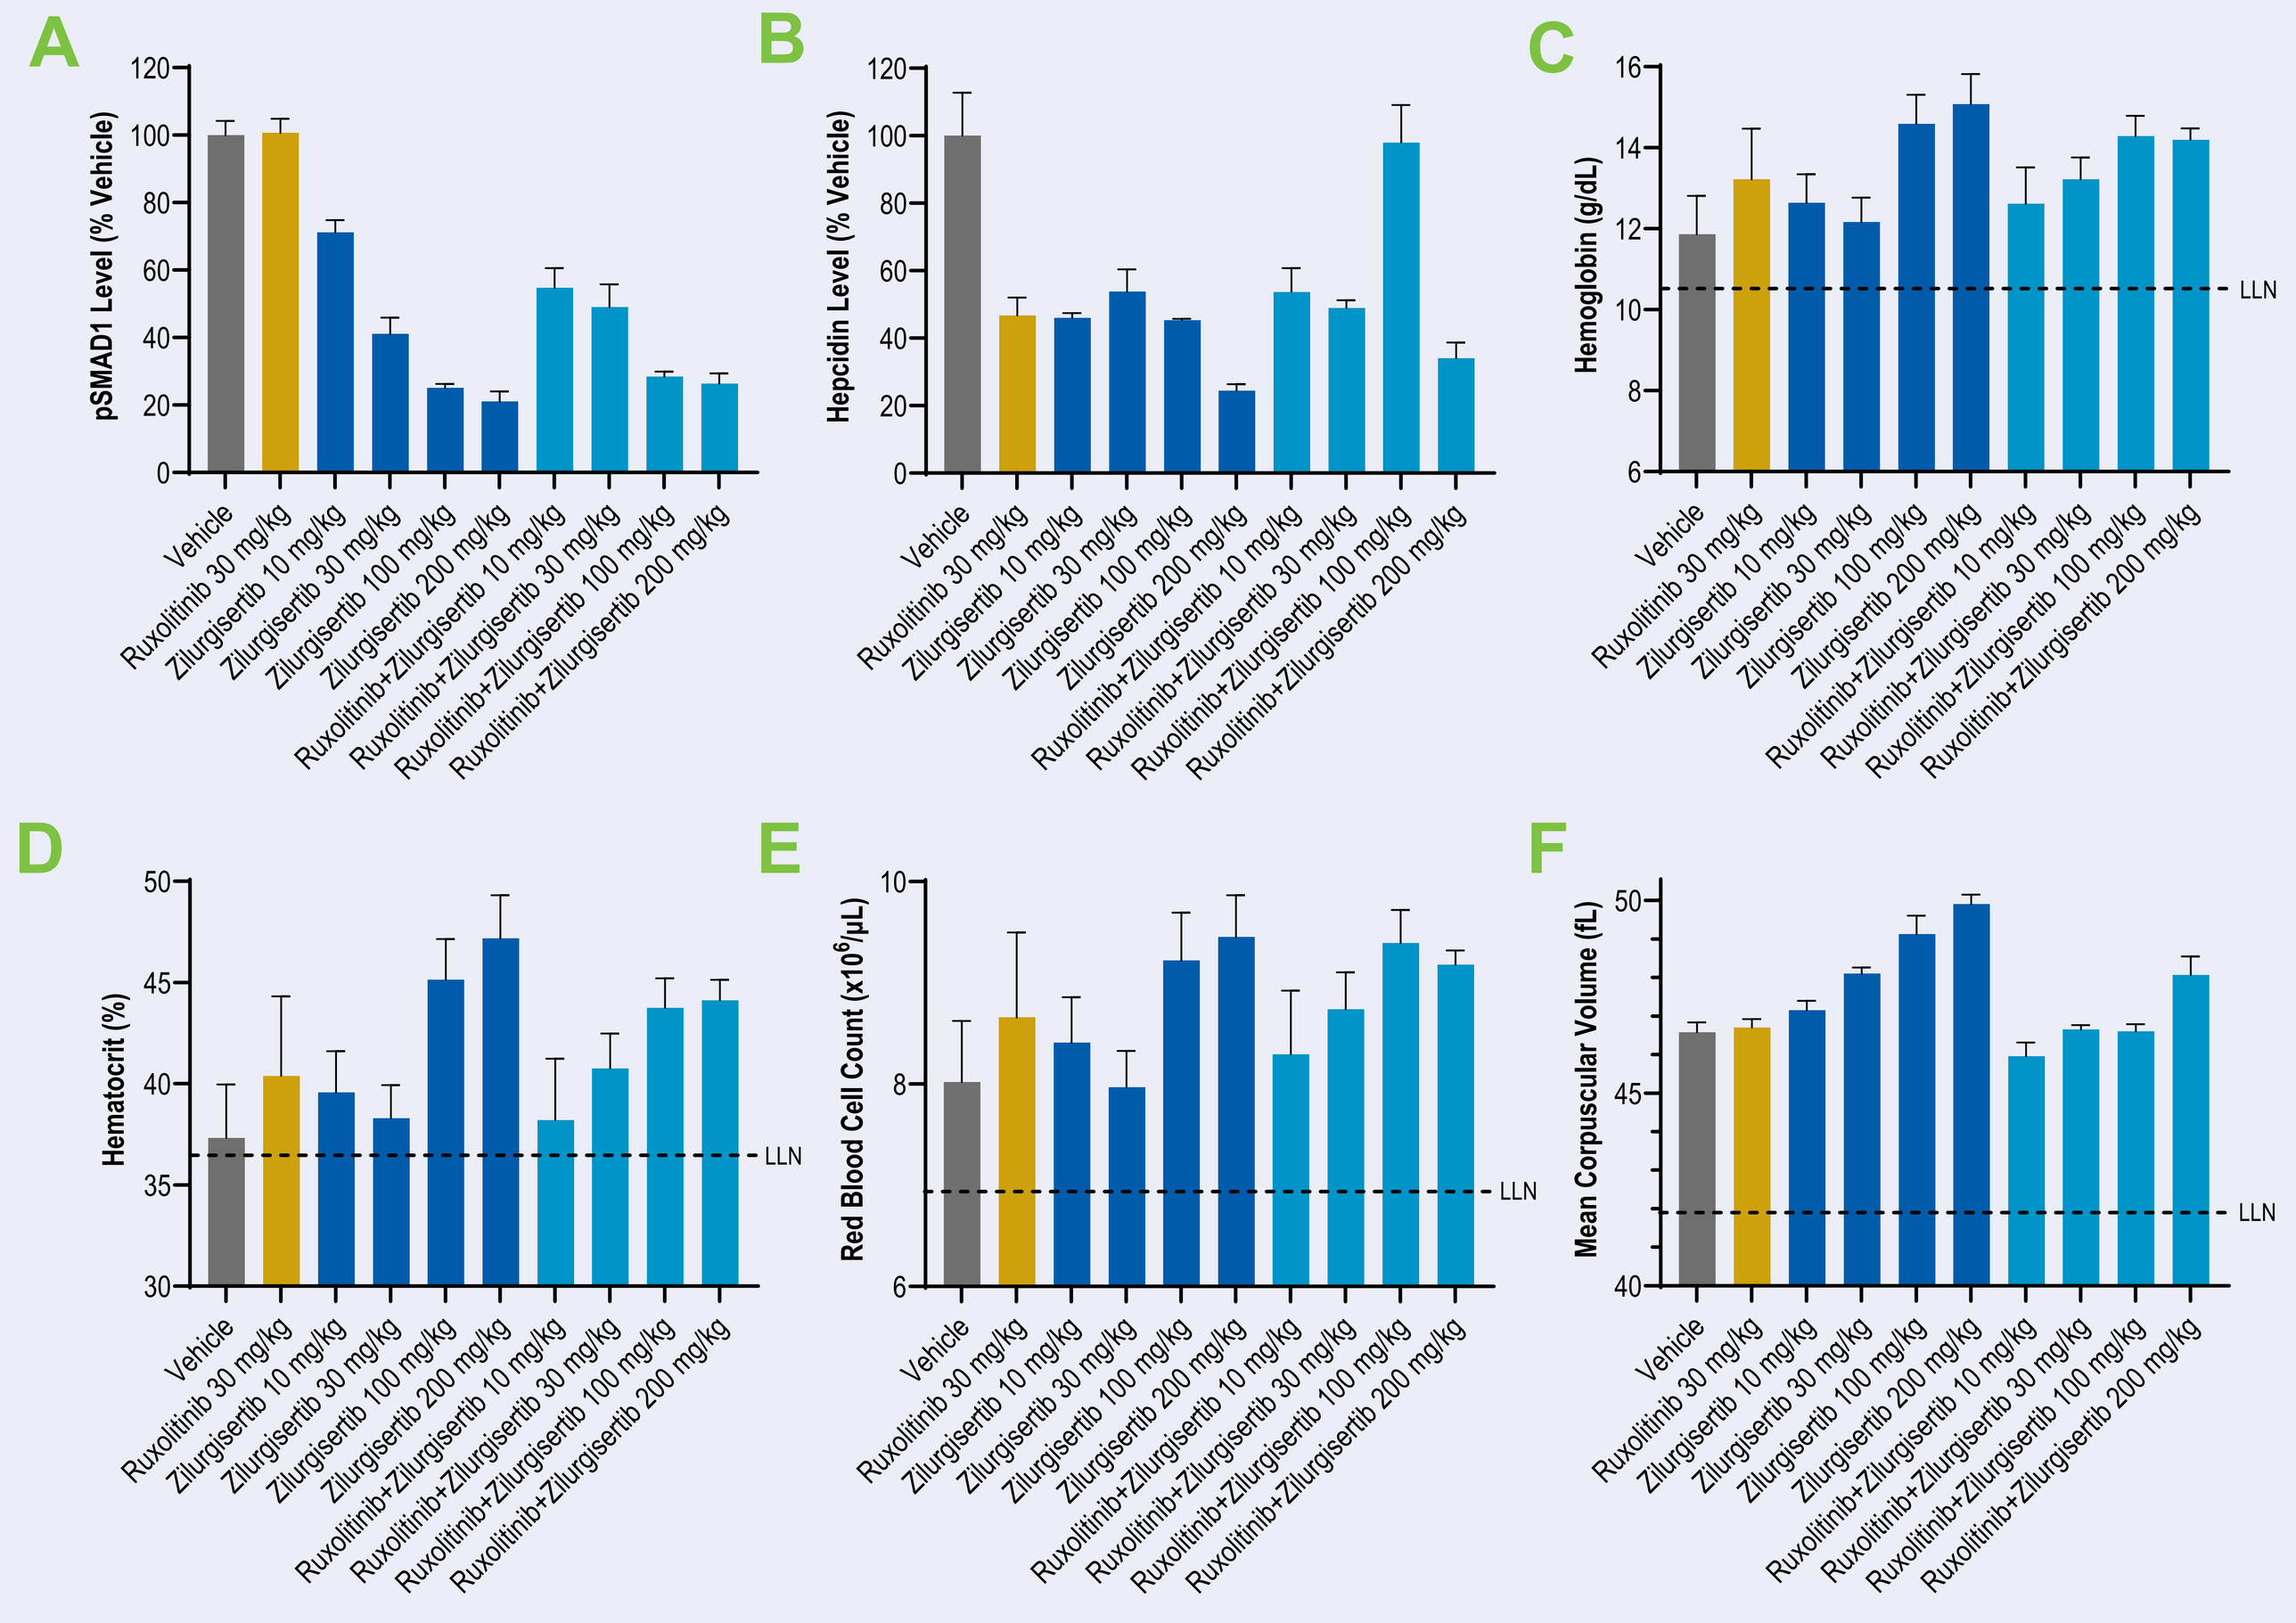
A
B
C
D
E
F
pSMAD1 Level (% Vehicle)
Hepcidin Level (% Vehicle)
Hemoglobin (g/dL)
Hematocrit (%)
Mean Corpuscular Volume (fL)
LLN
Vehicle
Ruxolitinib 30 mg/kg
Zilurgisertib 10 mg/kg
Zilurgisertib 30 mg/kg
Zilurgisertib 100 mg/kg
Zilurgisertib 200 mg/kg
Ruxolitinib+Zilurgisertib 10 mg/kg
Ruxolitinib+Zilurgisertib 30 mg/kg
Ruxolitinib+Zilurgisertib 100 mg/kg
Ruxolitinib+Zilurgisertib 200 mg/kg

Data represent means ± standard deviations. Ruxolitinib dosed at 30 mg/kg BID. BID, twice daily; LLN, lower limit of normal.

- B16F10 cells were injected intraperitoneally into female C57BL/6 mice. Two days after inoculation, mice were orally dosed for 7 days with vehicle, zilurgisertib, ruxolitinib, or a combination of the ALK2 inhibitors. Complete blood counts were then performed using a Sysmex XN analyzer **(A-F)**
- Hepcidin was analyzed from plasma samples using a murine-specific hepcidin enzyme-linked immunosorbent assay (ELISA) kit (Intrinsic LifeSciences, La Jolla, CA, USA) **(B)**
- At completion, liver samples were homogenized and lysates generated in Cell Lysis Buffer (Cell Signaling Technologies, Danvers, MA, USA); pSMAD1 levels were determined by ELISA (Cell Signaling Technologies) **(A)**
- Zilurgisertib dose dependently inhibits the ALK2/SMAD pathway, producing antianemic effects unaffected by ruxolitinib

Conclusions

- **Zilurgisertib is a potent and selective ALK2 inhibitor**
- **Biochemical and cell-based assays indicate that zilurgisertib directly inhibits ALK2 activity, reducing both phosphorylation of the direct target SMAD1, and levels of the SMAD target gene hepcidin required for iron homeostasis**
- **Zilurgisertib shows very little off-target activity, as determined by whole kinome profiling and by cell viability assays**
- **Zilurgisertib is efficacious in vivo:**
 - **Treatment with zilurgisertib inhibits the ALK2-SMAD1 pathway, resulting in reduced levels of hepcidin in a murine cancer-induced anemia model**
 - **Anemic mice treated with zilurgisertib show improved red blood cell parameters**
 - **The antianemia activity of zilurgisertib is unaffected by ruxolitinib**
- **Zilurgisertib in combination with ruxolitinib appears to be a rational and attractive approach to reduce anemia in patients with MF warranting further investigation**

Acknowledgments

The study was sponsored by Incyte Corporation (Wilmington, DE, USA). Editorial and graphics support was provided by Envision Pharma Group (Philadelphia, PA, USA) and funded by Incyte Corporation.

References

1. Verstovsek S, et al. *Lancet*. 2023;401:269-80. **2.** Oh S, et al. *Clin Lymphoma Myeloma Leuk*. 2022;22(S2):S237.